Subgroup Analysis of FOCUS Phase 3 Trial Efficacy Results.

1127P

Matthew Wheater[1], Marlana M. Orloff[2], Pier Francesco Ferrucci[3], Junsung Choi[4], David J. Eschelman[2], Evan S. Glazer[5], Aslam Ejaz[6], Erika Richtig[7], Sebastian Ochsenreither[8], Sunil A. Reddy[9], Michael C. Lowe[10], Georgia Beasley[11], Anja H. Gesierich[12], Martin Gschnell[13], Reinhard Dummer[14], Ana Maria Arance[15], Stephen W. Fenwick[16], Johnny John[17], Christian H.H. Ottensmeier[18], Jonathan S. Zager[4]

[1]University Hospital Southampton, Southampton, United Kingdom; [2]Thomas Jefferson University, Philadelphia, PA; [3]Gruppo Multimedica, Milan, Italy; [4]Moffitt Cancer Center, Tampa, FL; [5]University of Tennessee, Memphis, TN; [6]The Ohio State University, Columbus, OH; [7]Medical University of Graz, Graz, Austria; [8]Charité Hospital Berlin, Berlin, Germany; [9]Stanford University, Stanford, CA; [10]Emory University, Atlanta, GA; [11]Duke University, Durham, NC; [12]University Hospital Würzburg, Würzburg, Germany; [13]University Hospital Giessen and Marburg, Marburg, Germany; [14]University Hospital Zürich, Zürich, Switzerland; [15]Hospital Clinic of Barcelona, Barcelona, Spain; [16]Liverpool University Hospitals, Liverpool, United Kingdom; [17]Delcath Systems, Queensbury, NY; [18]University of Liverpool, Liverpool, United Kingdom

Background

- Metastatic uveal melanoma (mUM) has a poor prognosis, with liver metastases typically presenting a therapeutic challenge.[1]
- Liver metastasis is the most common cause of death for patients with mUM.[2]
- Melphalan/Hepatic Delivery System (melphalan/HDS) is a drug/device combination used in the percutaneous hepatic perfusion (PHP) procedure for liver-directed treatment of unresectable metastatic tumors in mUM patients.
- The PHP procedure uniquely treats the entire liver by isolating liver circulation, saturating the entire liver with a high dose of melphalan, and then filtering the blood extracorporeally to remove up to 85% of the administered melphalan prior to returning the blood to systemic circulation.

ISOLATION

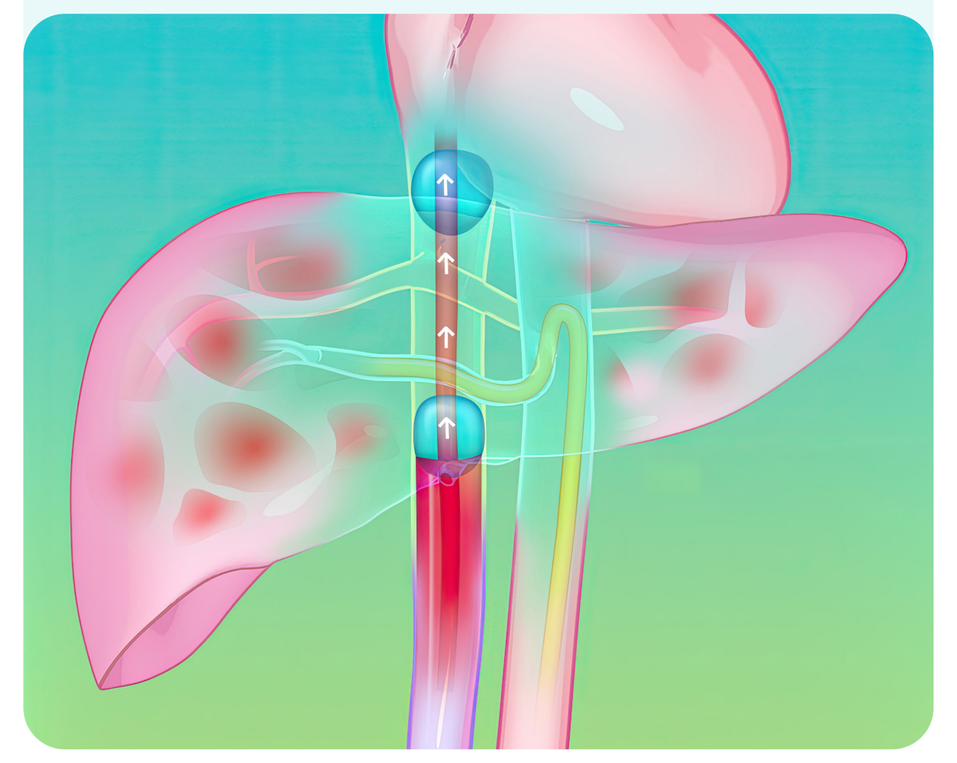

Liver isolated via Double Balloon Catheter in Inferior Vena Cava

SATURATION

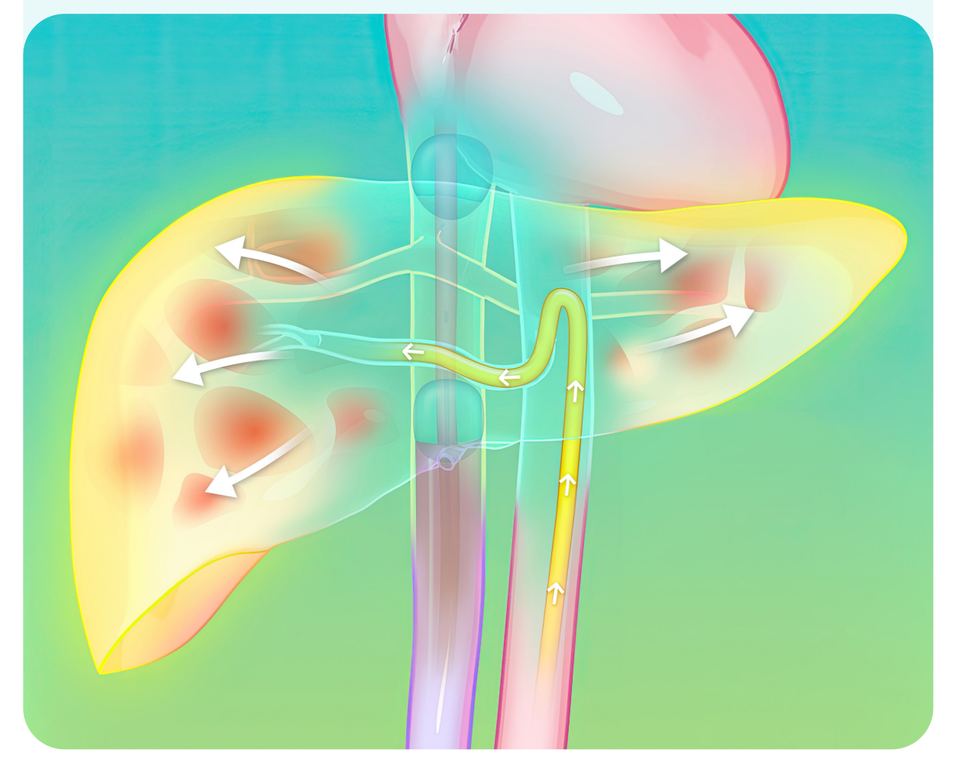

Melphalan infused directly into liver via catheter in Hepatic Artery

FILTRATION

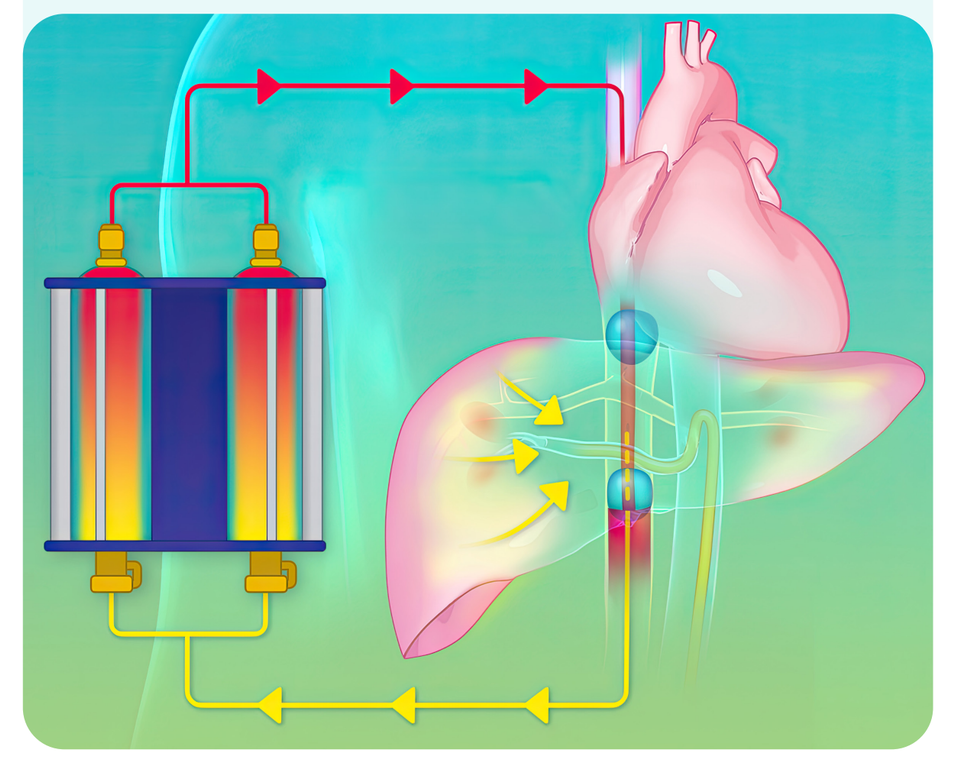

Blood exiting the liver filtered by Extracorporeal Filters

Methods

- The FOCUS Trial treated 91 patients with melphalan/HDS in the US and Europe.
- Patients were treated with melphalan at 3 mg/kg ideal body weight (maximum dose: 220 mg per treatment) every 6-8 weeks for up to 6 cycles.
- Tumor response was assessed by CT or MRI every 12 (±2) weeks using RECIST 1.1 criteria.
- Patients with hepatic or extrahepatic progressive disease (PD) were discontinued from study treatment. All patients were followed until death.
- Efficacy endpoints including objective response rate (ORR), progression-free survival (PFS), overall survival (OS), were assessed for subgroups of patients with and without extrahepatic disease, previously treated and treatment-naïve patients, and those with low and higher extent of liver involvement.
- Onset of tumor response and rates of serious adverse events (SAEs) and Grade 3/4 adverse events (AE) were assessed by treatment cycle.

Key Inclusion Criteria

- 50% or less liver involvement from metastatic uveal melanoma.
- Liver disease must be measurable by CT and/or MRI.
- Limited extrahepatic disease at baseline permitted if life-threatening component of disease is in liver.
- ECOG performance status of 0-1 at screening.
- Prior chemotherapy, radiotherapy, chemoembolization, radioembolization, or immunoembolization permitted after washout period of 30 days.
- Prior PD-1 immunotherapy, such as pembrolizumab or nivolumab, or anti–CTLA-4 immunotherapy, such as ipilimumab, permitted after washout period of 8 weeks.

Key Exclusion Criteria

- Child-Pugh Class B or C cirrhosis or evidence of portal hypertension.
- New York Heart Association functional classification II, III or IV active cardiac conditions, or any cardiac conditions precluding use of general anesthesia.
- Clinically significant pulmonary disease that precludes use of general anesthesia.
- Prior Whipple procedure.
- Patients on immunosuppressive drugs or who cannot be temporarily removed from chronic anticoagulation therapy
- Patients with active bacterial infections with systemic manifestations (eg, malaise, fever, leukocytosis) are not eligible until completion of appropriate therapy.

REFERENCES

1. Kaštelan S, et al. Front Biosci (Landmark Ed). 2022 Feb 21;27(2):72.
2. Bakalian S, et al. Clin Cancer Res. 2008;14(4):951-956.

SCAN TO LEARN MORE

Key Results

- Significant differences in OS were observed based on extent of liver involvement.
- There were no significant differences in OS based on presence of extrahepatic lesions or prior therapy.
- There were no significant differences in ORR or PFS based on presence of extrahepatic lesions, prior therapy, or extent of liver involvement.
- Rates of SAEs and Grade 3/4 AEs were largely consistent across treatment cycles, suggesting an absence of cumulative toxicities after multiple treatments.

Figure 1: PFS by Presence of Extrahepatic Lesions

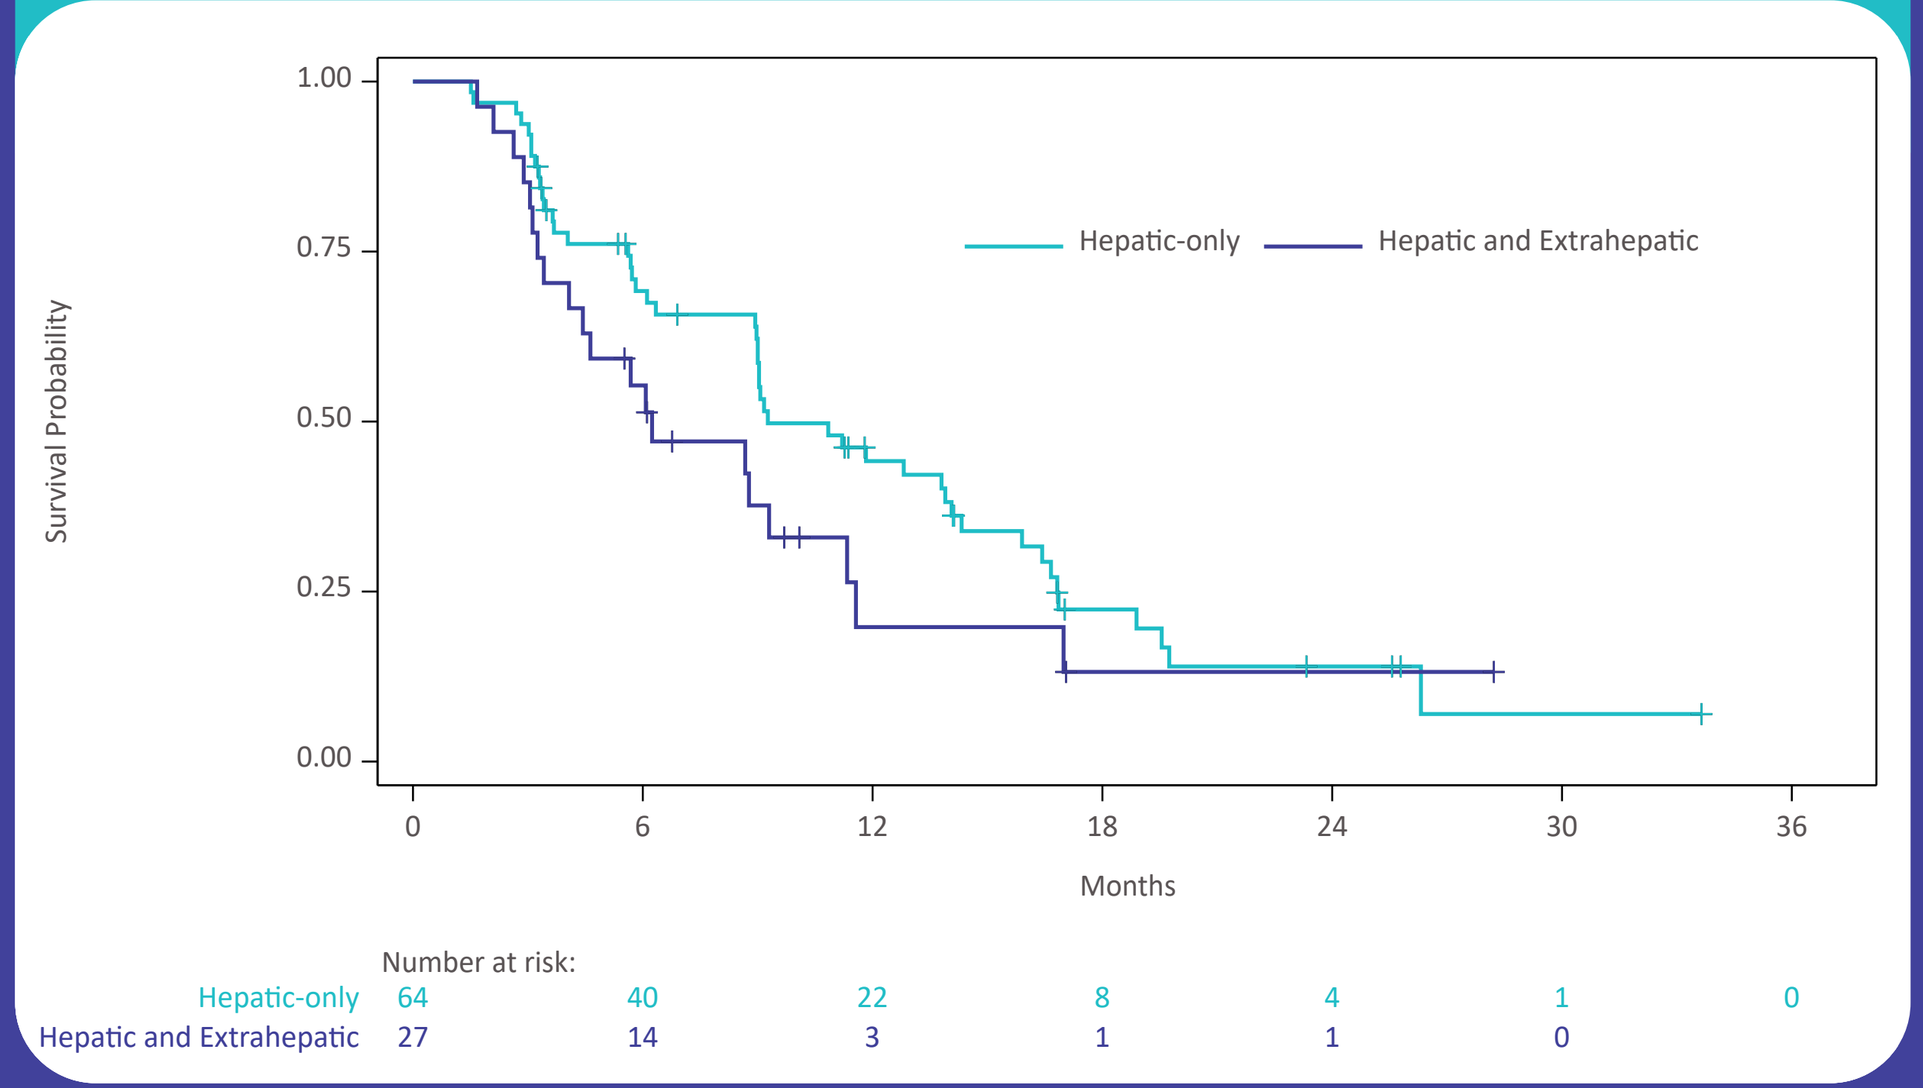
Survival Probability
1.00
0.75
0.50
0.25
0.00
Hepatic-only
Hepatic and Extrahepatic
0 6 12 18 24 30 36
Months
Number at risk:
Hepatic-only 64 40 22 8 4 1 0
Hepatic and Extrahepatic 27 14 3 1 1 0

Figure 2: OS by Presence of Extrahepatic Lesions

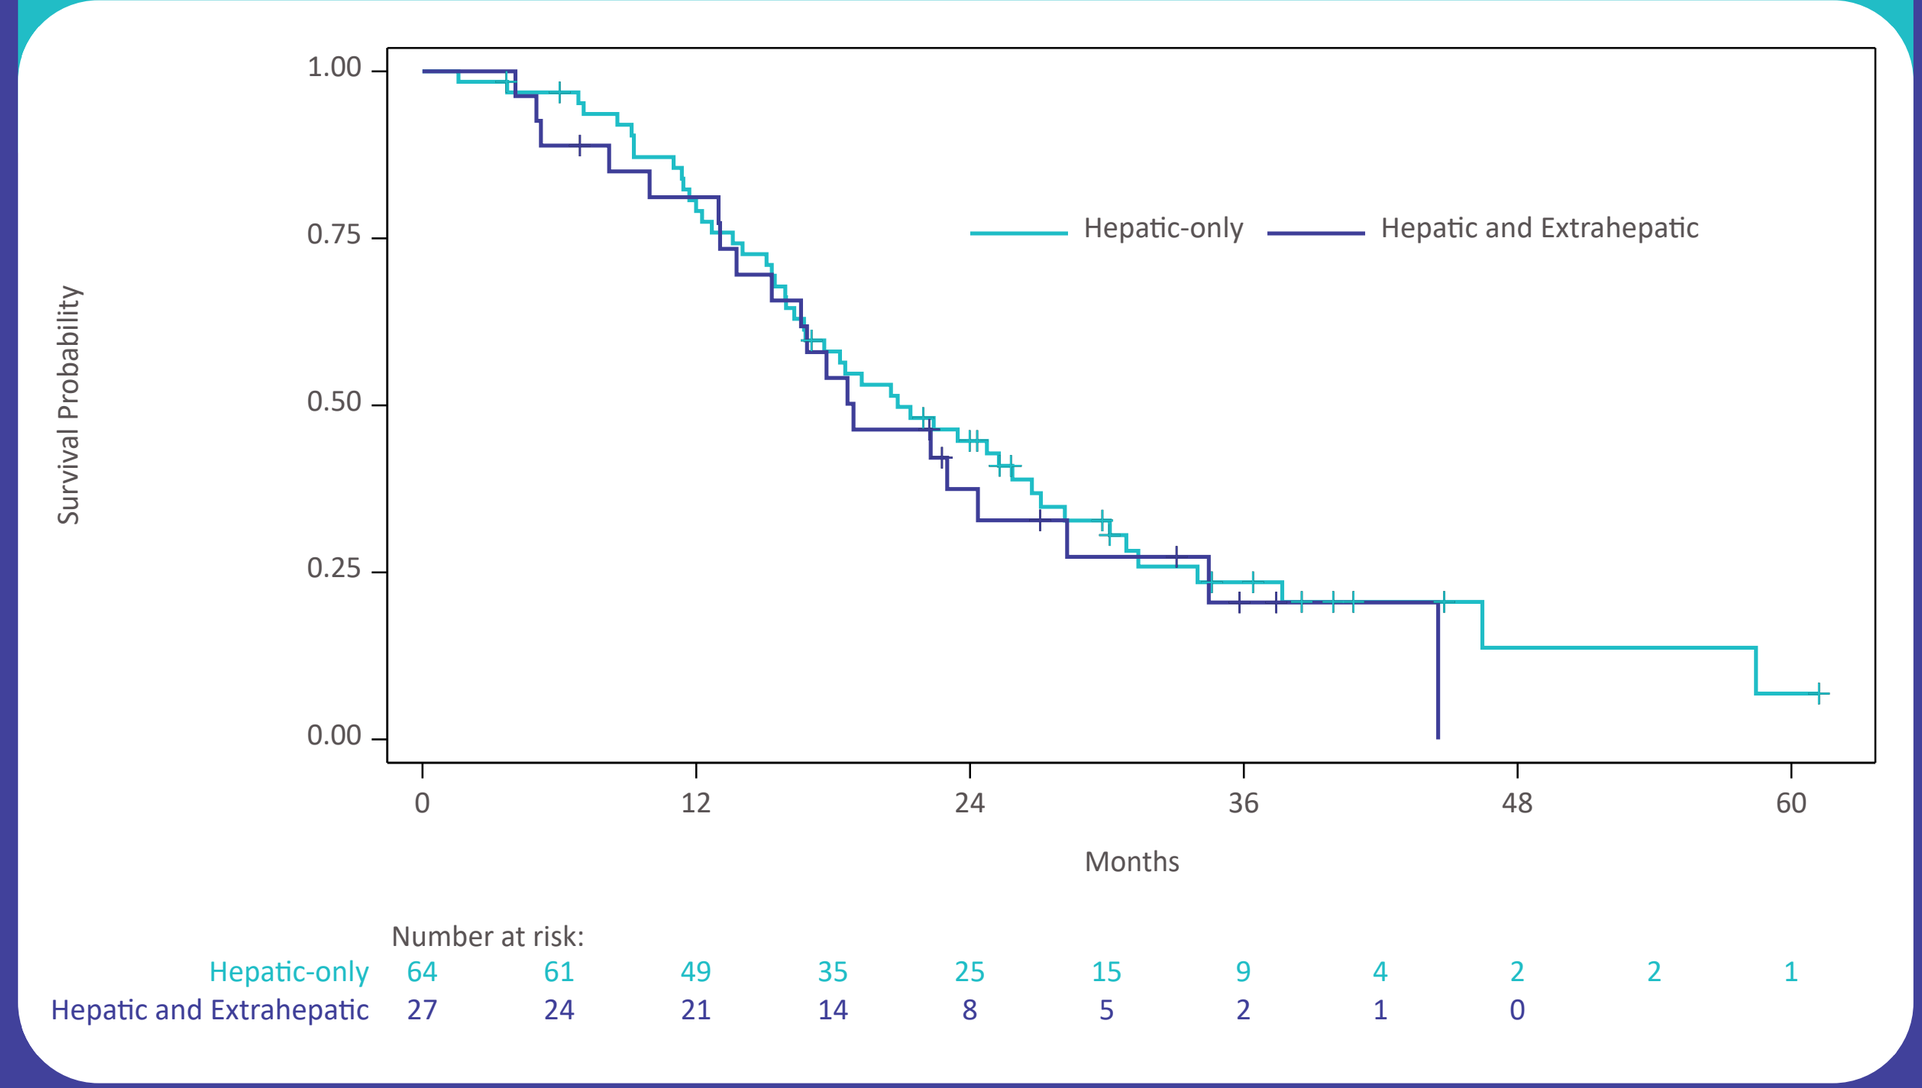
Survival Probability
1.00
0.75
0.50
0.25
0.00
Hepatic-only
Hepatic and Extrahepatic
0 12 24 36 48 60
Months
Number at risk:
Hepatic-only 64 61 49 35 25 15 9 4 2 2 1
Hepatic and Extrahepatic 27 24 21 14 8 5 2 1 0

Figure 3: PFS by Prior Therapy

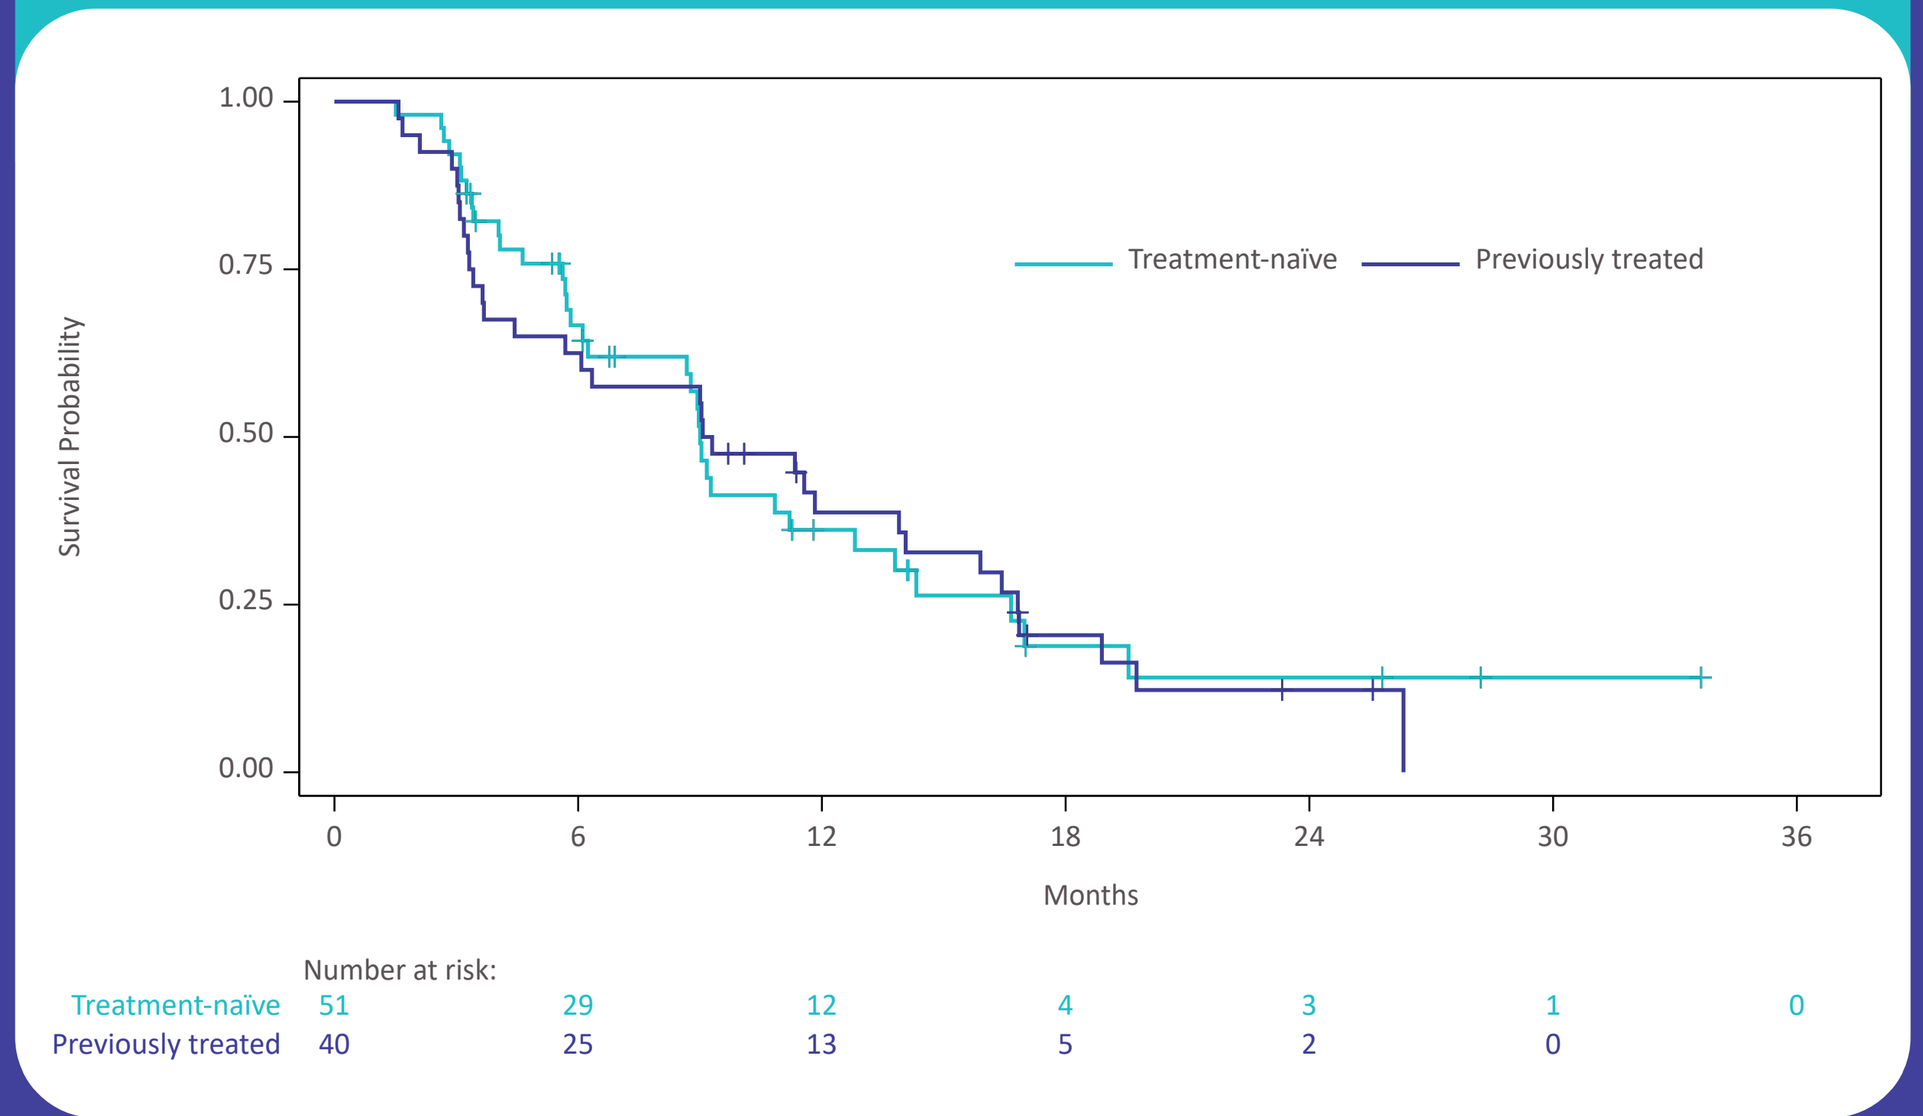
Survival Probability
1.00
0.75
0.50
0.25
0.00
Treatment-naïve
Previously treated
0 6 12 18 24 30 36
Months
Number at risk:
Treatment-naïve 51 29 12 4 3 1 0
Previously treated 40 25 13 5 2 0

Figure 4: OS by Prior Therapy

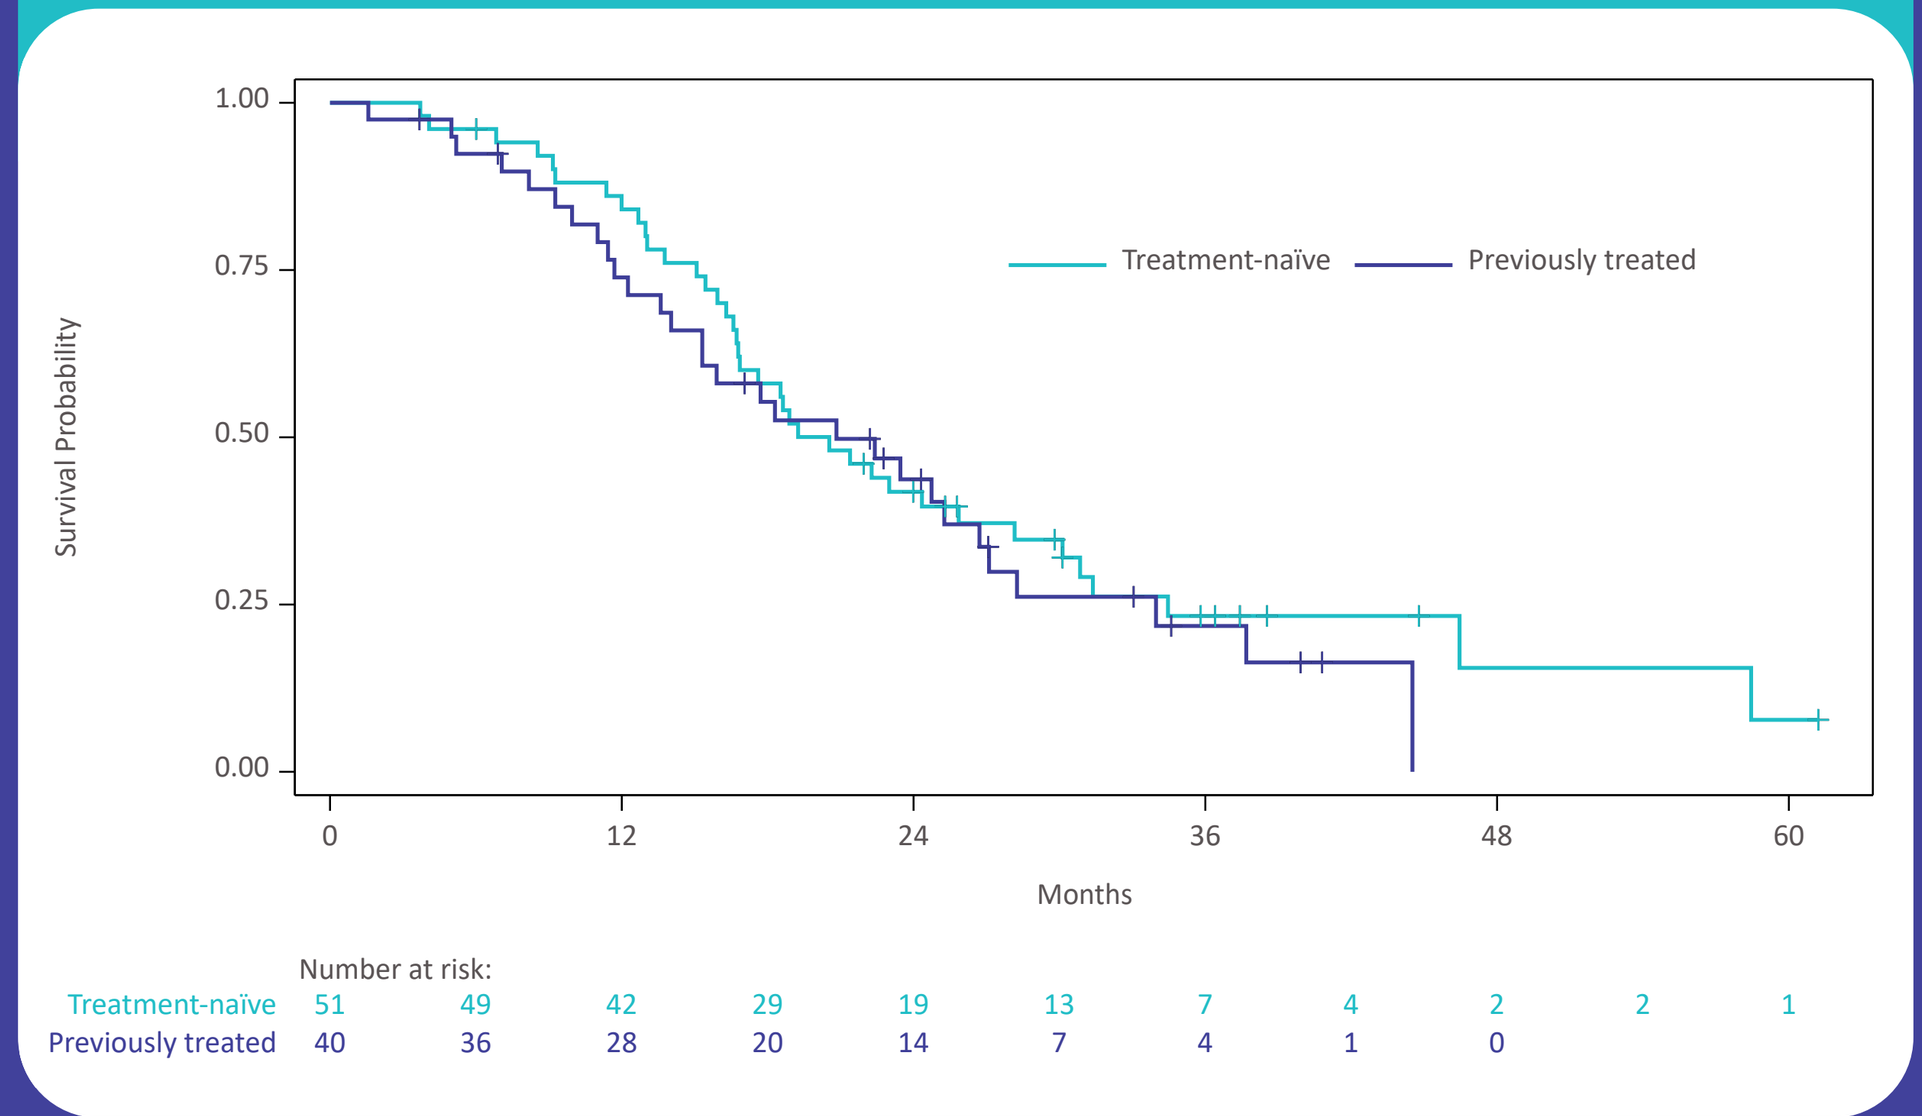
Survival Probability
1.00
0.75
0.50
0.25
0.00
Treatment-naïve
Previously treated
0 12 24 36 48 60
Months
Number at risk:
Treatment-naïve 51 49 42 29 19 13 7 4 2 2 1
Previously treated 40 36 28 20 14 7 4 1 0

Figure 5: PFS by Extent of Liver Involvement

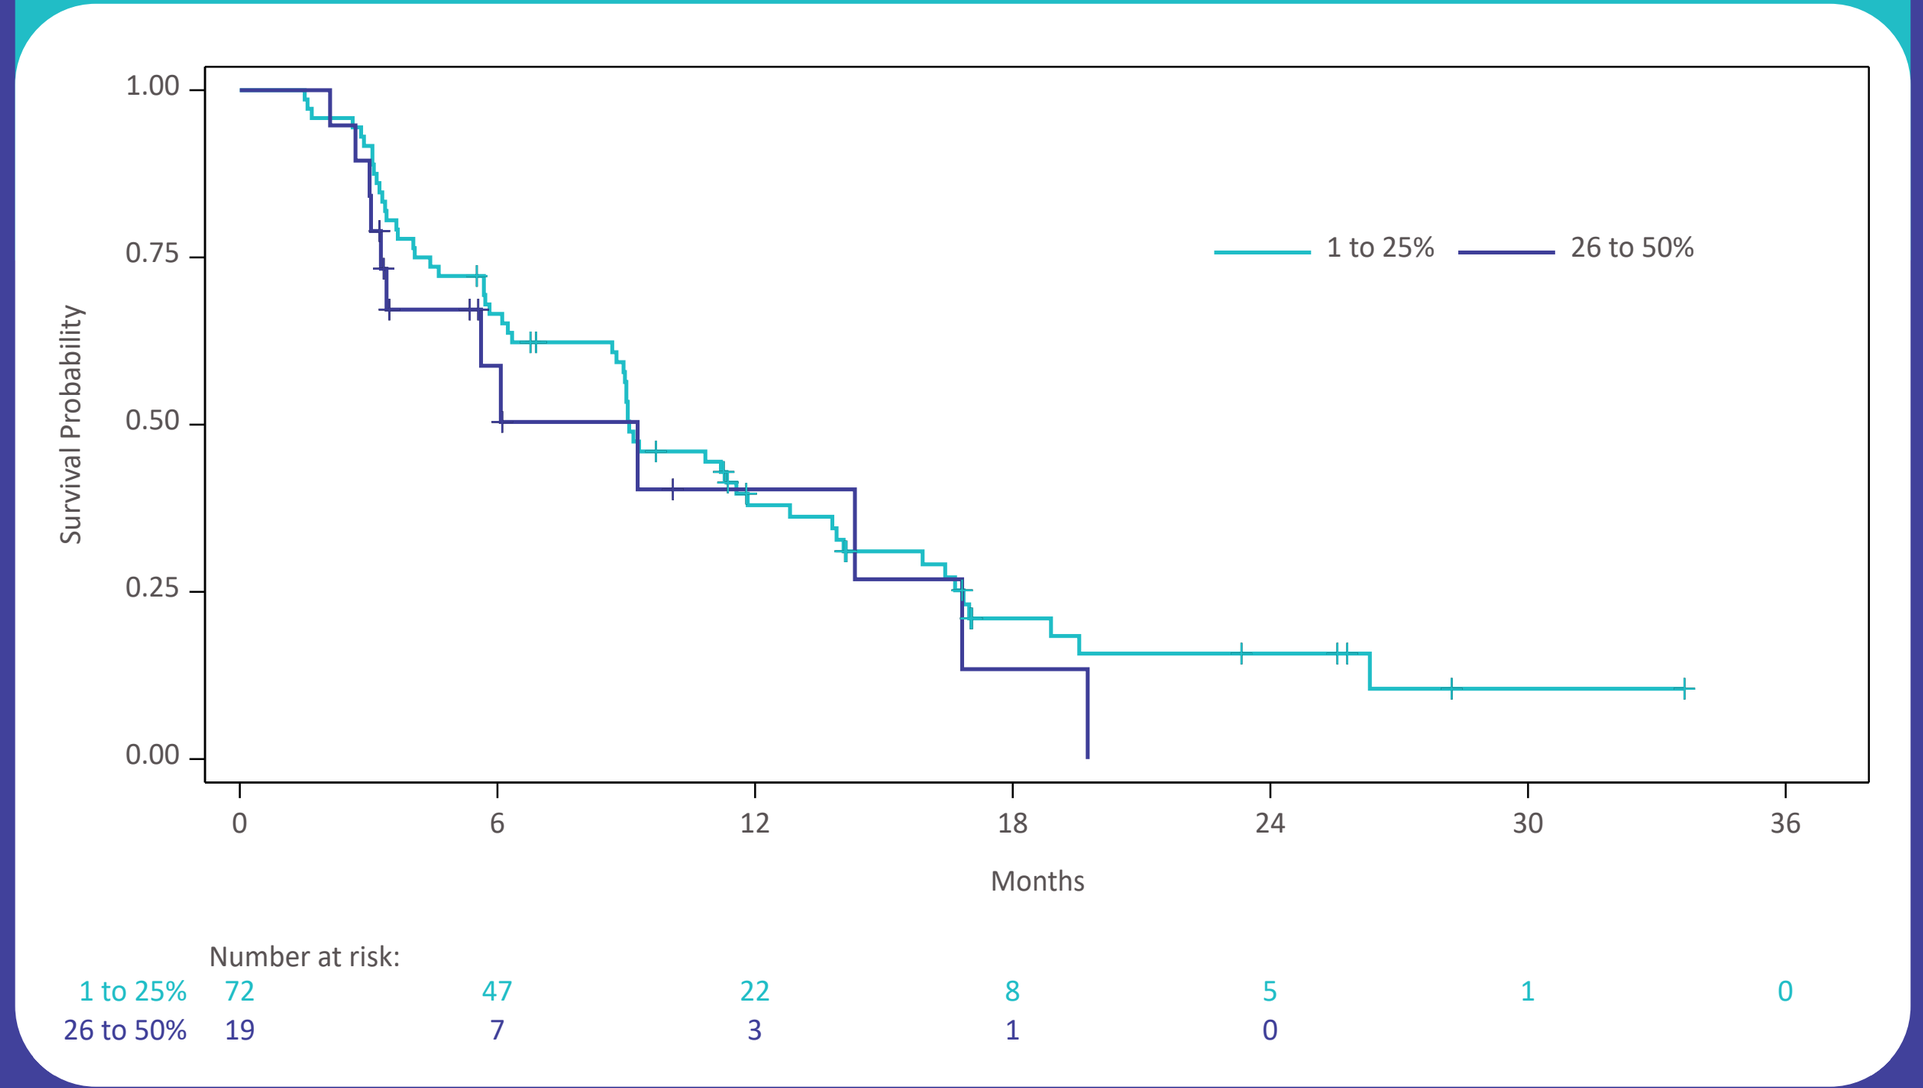
Survival Probability
1.00
0.75
0.50
0.25
0.00
1 to 25%
26 to 50%
0 6 12 18 24 30 36
Months
Number at risk:
1 to 25% 72 47 22 8 5 1 0
26 to 50% 19 7 3 1 0

Figure 6: OS by Extent of Liver Involvement

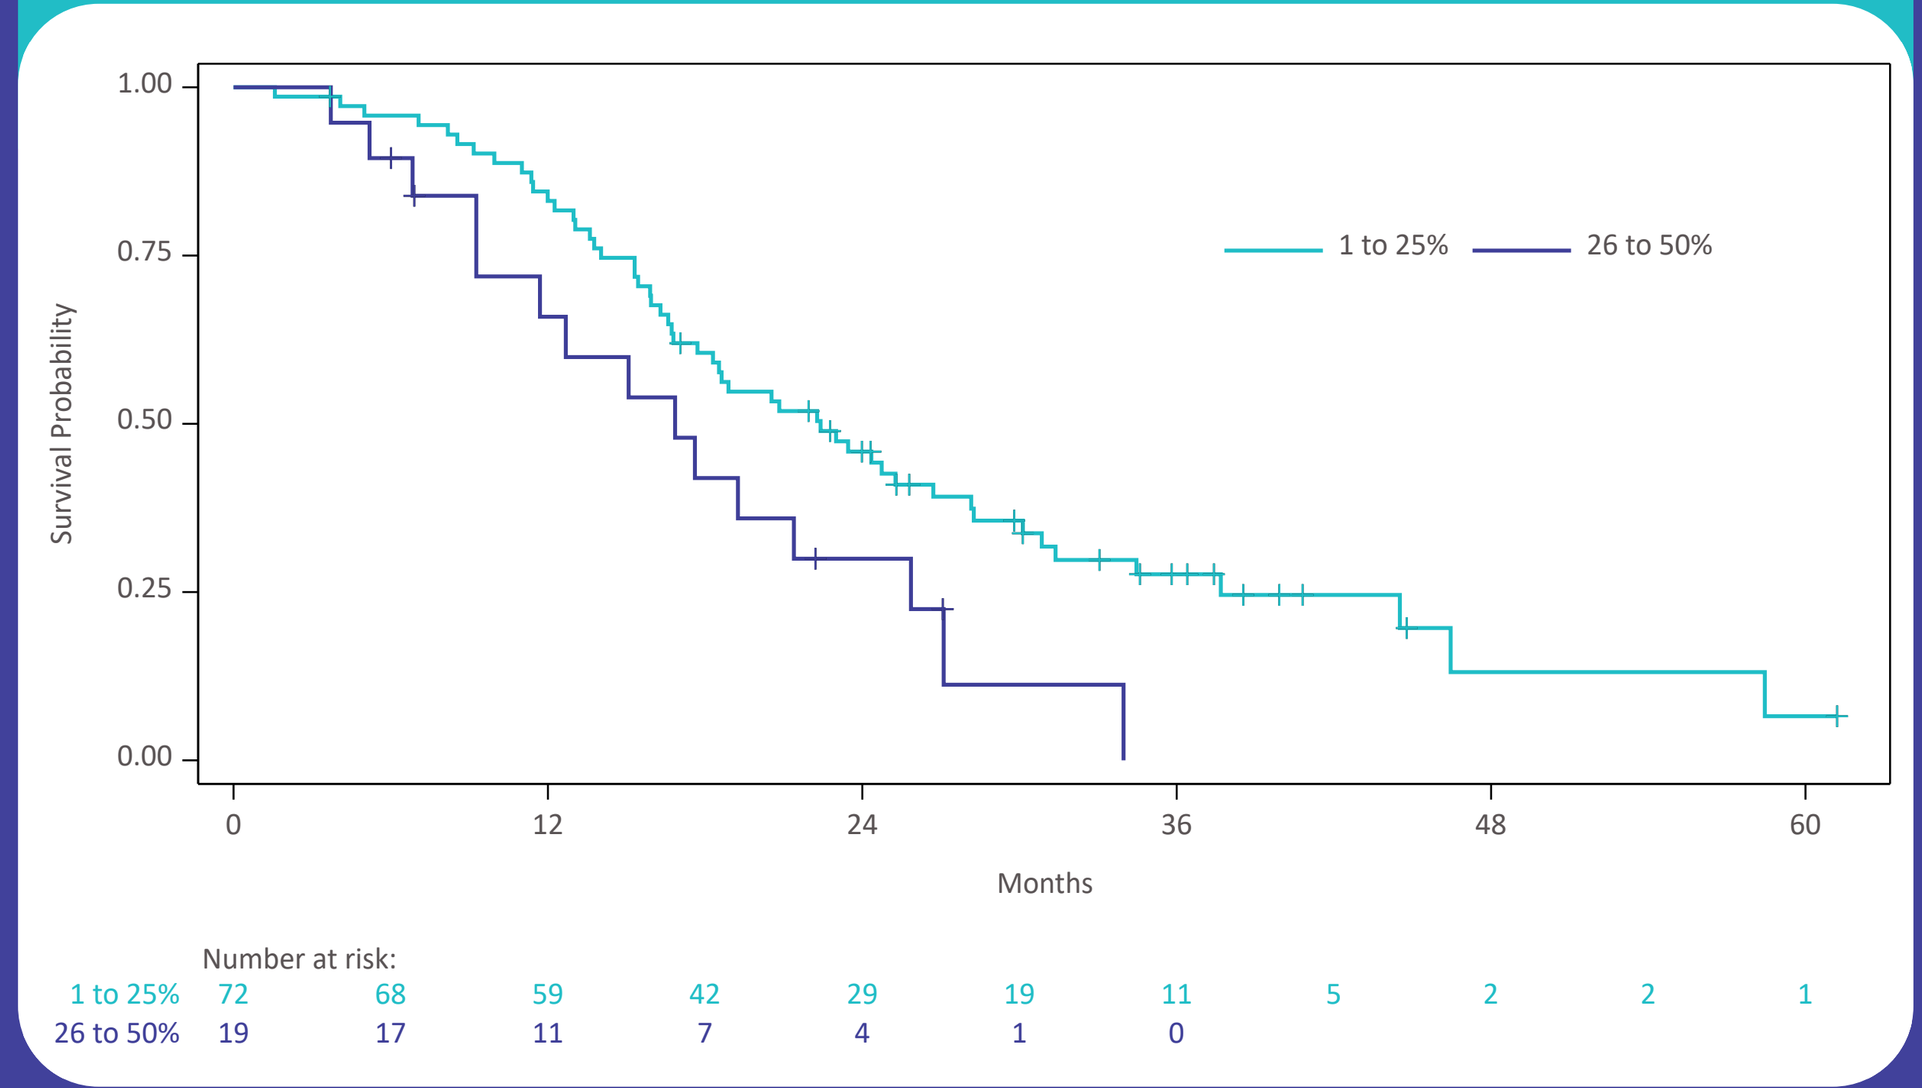
Survival Probability
1.00
0.75
0.50
0.25
0.00
1 to 25%
26 to 50%
0 12 24 36 48 60
Months
Number at risk:
1 to 25% 72 68 59 42 29 19 11 5 2 2 1
26 to 50% 19 17 11 7 4 1 0

Detailed Results

Table 1. Baseline Characteristics by Subgroup (Treated population)

Characteristic	Melphalan/HDS (N = 91)
Age group — n (%)	
< 65 years	61 (67.0)
≥ 65 years	30 (33.0)
Gender — n (%)	
Male	44 (48.4)
Female	47 (51.6)
Lactate dehydrogenase — n/N (%)	
Low or normal	54/86 (62.8)
Elevated	32/86 (37.2)
Geographic region — n (%)	
Europe	45 (49.5)
United States	46 (50.5)
Presence of extrahepatic lesions — n (%)[1]	
Hepatic only	64 (70.3)
Hepatic and extrahepatic[2]	27 (29.7)
Prior therapy — n (%)	
Treatment-naïve	51 (56.0)
Previously treated	40 (44.0)
Extent of liver involvement — n (%)[3]	
1-25%	72 (79.1)
26–50%	19 (20.9)

[1] Based on Independent Review Committee assessment.
[2] Extrahepatic lesions include lung, lymph node, bone (spine, lumbar spine, pelvis, ribs, sacrum, and skull), soft tissue (subcutaneous, trunk, and chest wall), and other visceral (spleen and adrenal gland).
[3] Assessed by the investigator.

Table 2. AEs by Subgroup (Safety population)

Characteristic	N	SAE n (%)	Grade 3/4 AE n (%)
Overall	95	43 (45.3)	77 (81.1)
Age group			
< 65 years	65	30 (46.2)	53 (81.5)
≥ 65 years	30	13 (43.3)	24 (80.0)
Gender			
Male	47	22 (46.8)	35 (74.5)
Female	48	21 (43.8)	42 (87.5)
LDH			
Low or normal	55	28 (50.9)	48 (87.3)
Elevated	35	13 (37.1)	25 (71.4)
Geographic region			
Europe	46	20 (43.5)	35 (76.1)
United States	49	23 (46.9)	42 (85.7)
Presence of extrahepatic lesions[1]			
Hepatic only	66	35 (53.0)	55 (83.3)
Hepatic and extrahepatic[2]	27	7 (25.9)	21 (77.8)
Prior therapy			
Treatment-naïve	54	23 (42.6)	41 (75.9)
Previously treated	41	20 (48.8)	36 (87.8)
Extent of liver involvement[3]			
1-25%	75	33 (44.0)	60 (80.0)
26–50%	20	10 (50.0)	17 (85.0)

Table 3. Adverse Events by Treatment Cycle (Treated Population)

Treatment Cycle	SAE n (%)	Grade 3/4 AE n (%)
Cycle 1 (N=91)	20 (22.0)	49 (53.8)
Cycle 2 (N=84)	13 (15.5)	48 (57.1)
Cycle 3 (N=66)	9 (13.6)	35 (53.0)
Cycle 4 (N=55)	3 (5.5)	25 (45.5)
Cycle 5 (N=40)	3 (7.5)	20 (50.0)
Cycle 6 (N=34)	6 (17.6)	16 (47.1)

Table 4. Objective Response by Treatment Cycle (Treated population - Assessed by IRC)

Treatment Cycle of First Objective Response	Patients with Objective Response (N = 33) n (%)
Cycle 1	3 (9.1)
Cycle 2	16 (48.5)
Cycle 3	3 (9.1)
Cycle 4	8 (24.2)
Cycle 5	1 (3.0)
Cycle 6	2 (6.1)

Table 5. Efficacy Results by Subgroup

	Presence of Extrahepatic Lesions		Prior Therapy		Extent of Liver Involvement	
	Hepatic only (n = 64)	Hepatic and extrahepatic (n = 27)	Treatment-naïve (n = 51)	Previously treated (n = 40)	1 to 25% (n = 72)	26 to 50% (n = 19)
Objective Response Rate, ORR						
% (n)	37.5 (24)	33.3 (9)	35.3 (18)	37.5 (15)	37.5 (27)	31.6 (6)
95% CI[a]	25.70-50.49	16.52-53.96	22.43-49.93	22.73-54.20	26.36-49.70	12.58-56.55
P Value[b]	.8131		.8302		.7901	
[a] Exact binomial CI. [b] Fisher exact test.						
Best Overall Response, BOR, n (%)						
Complete response, CR	6 (9.4)	1 (3.7)	3 (5.9)	4 (10.0)	7 (9.7)	0
Partial response, PR	18 (28.1)	8 (29.6)	15 (29.4)	11 (27.5)	20 (27.8)	6 (31.6)
Stable disease, SD	25 (39.1)	9 (33.3)	23 (45.1)	11 (27.5)	27 (37.5)	7 (36.8)
Progressive Disease, PD	14 (21.9)	9 (33.3)	10 (19.6)	13 (32.5)	17 (23.6)	6 (31.6)
Not evaluable, NE	1 (1.6)	0	0	1 (2.5)	1 (1.4)	0
Progression-Free Survival, PFS						
Events, n (%)	47 (73.4)	20 (74.1)	34 (66.7)	33 (82.5)	55 (76.4)	12 (63.2)
Censored, n (%)	17 (26.6)	7 (25.9)	17 (33.3)	7 (17.5)	17 (23.6)	7 (36.8)
Median (95% CI), months	9.26 (8.97-14.06)	6.24 (3.42-11.33)	9.00 (6.11-12.81)	9.18 (4.44-14.06)	9.07 (8.67-11.83)	9.26 (3.29-16.82)
P Value[a]	.1642		.8598		.3767	
Hazard ratio (95% CI)	0.71 (0.42-1.21)		0.93 (0.57-1.52)		0.74 (0.40-1.40)	
P Value	.2085		.7773		.3587	
At 6 months						
Patients alive and without progression, n (%)	40 (62.5)	14 (51.9)	29 (56.9)	25 (62.5)	47 (65.3)	7 (36.8)
At 12 months						
Patients alive and without progression, n (%)	22 (34.4)	3 (11.1)	12 (23.5)	13 (32.5)	22 (30.6)	3 (15.8)
[a] Log-rank test.						
Overall Survival, OS						
Events, n (%)	47 (73.4)	20 (74.1)	38 (74.5)	29 (72.5)	52 (72.2)	15 (78.9)
Censored, n (%)	17 (26.6)	7 (25.9)	13 (25.5)	11 (27.5)	20 (27.8)	4 (21.1)
Median (95% CI), months	20.83 (16.30-26.71)	18.89 (13.77-28.25)	20.53 (16.72-28.16)	20.83 (14.03-26.71)	22.41 (16.79-28.16)	16.85 (9.26-25.86)
P Value[a]	.5931		.4988		.0296	
Hazard ratio (95% CI)	0.88 (0.52-1.48)		0.88 (0.54-1.44)		0.53 (0.29-0.95)	
P Value	.6208		.6071		.0325	
At 12 months						
Patients alive, n (%)	49 (76.6)	21 (77.8)	42 (82.4)	28 (70.0)	59 (81.9)	11 (57.9)
At 24 months						
Patients alive, n (%)	25 (39.1)	8 (29.6)	19 (37.3)	14 (35.0)	29 (40.3)	4 (21.1)
[a] Log-rank test.						

Conclusions

- Treatment with melphalan/HDS provides clinically meaningful efficacy across the evaluated subgroups.
- Objective tumor responses occurred throughout all 6 treatment cycles, without evidence of cumulative toxicity, demonstrating a favorable benefit-risk profile in patients who typically have a poor prognosis and limited treatment options.

For questions/comments, contact
Dr. Matthew Wheater at matthew.wheater@uhs.nhs.uk

Dr. Wheater received reimbursement from Delcath Systems for speaking engagements.

Presented at the ESMO Congress 2024 - European Society for Medical Oncology, 13-17 September 2024 in Barcelona, Spain.

Funding for the FOCUS Trial was provided by Delcath Systems.

AMER-M-XXXX (v1)